HIV-1 Resistance Analysis of Treatment-Naïve People With HIV and HBV Receiving B/F/TAF or DTG + F/TDF

Michelle L D'Antoni[1], **Archana V Boopathy**[1], **Kristen Andreatta**[1], **Silvia Chang**[1], **Jason T Hindman**[1], **Anchalee Avihingsanon**[2], **Laurie A VanderVeen**[1], **Christian Callebaut**[1]

[1]Gilead Sciences, Inc., Foster City, CA, USA; [2]HIV-NAT, Thai Red Cross AIDS Research Centre, Bangkok, Thailand

735

ALLIANCE

Conclusions

- In people with HIV-1 and HBV receiving DTG + F/TDF or B/F/TAF, preexisting RAMs were uncommon and did not affect HIV-1 virologic suppression through EOS
- Among participants meeting criteria for inclusion in the final RAP, none developed resistance to the study drugs
- Two emergent RAMs occurred in one participant who experienced nonadherence while receiving DTG + F/TDF, highlighting the importance of adherence in suppressing viral load and preventing treatment-emergent resistance
- These results provide further evidence of the long-term efficacy and high barrier to resistance associated with B/F/TAF

Plain Language Summary

- The ALLIANCE study looked at how well two treatments called B/F/TAF and DTG + F/TDF work to treat adults who had both human immunodeficiency virus type 1 (HIV-1) and hepatitis B virus (HBV) infections
 - After 96 weeks, both treatments lowered the levels of HIV-1 and HBV in the blood
- This analysis of the ALLIANCE study looks at genetic changes in HIV. These changes occur by chance and some of them stop HIV medicines from working; these changes are called resistance mutations
- People were tested at the start of the study to see if they had HIV resistance mutations
 - The results showed that most people did not have resistance mutations
 - When resistance mutations were present, they did not affect the ability of the treatments to lower the level of HIV-1 in the blood
- Researchers wanted to see if people with increases of HIV-1 in their blood during the study had developed resistance mutations during the study
 - One person, taking DTG + F/TDF, developed resistance mutations

Introduction

- Globally, approximately 3.1 million people are living with both HIV-1 and hepatitis B virus (HBV)[1]
- Tenofovir alafenamide (TAF)– or tenofovir disoproxil fumarate (TDF)–based antiretroviral therapy is recommended as an initial treatment for most adults and adolescents with HIV-1 and HBV[2-4]
- The ALLIANCE study demonstrated that bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF) was noninferior to dolutegravir (DTG) + F/TDF at achieving HIV-1 RNA suppression, and superior at achieving HBV DNA suppression, at Week 48 in treatment-naïve adults with both HIV-1 and HBV infections, with high rates of HIV-1 and HBV suppression also observed at Week 96[5]
- Resistance-associated mutations (RAMs) can reduce the effectiveness of HIV-1 treatments; guidelines recommend genotypic testing when starting treatment or during treatment failure, and that treatment options with a high barrier to resistance should be selected when possible[2,3,6,7]

Objective

- To report HIV-1 resistance analyses to the end of the ALLIANCE study up to ≥ 144 weeks, including the 48-week open-label extension (OLE) phase in which participants received B/F/TAF

Methods

- ALLIANCE (NCT03547908) was a randomized, double-blind, active-controlled, Phase 3 clinical study, which included a 48-week OLE phase[1]

Study Design

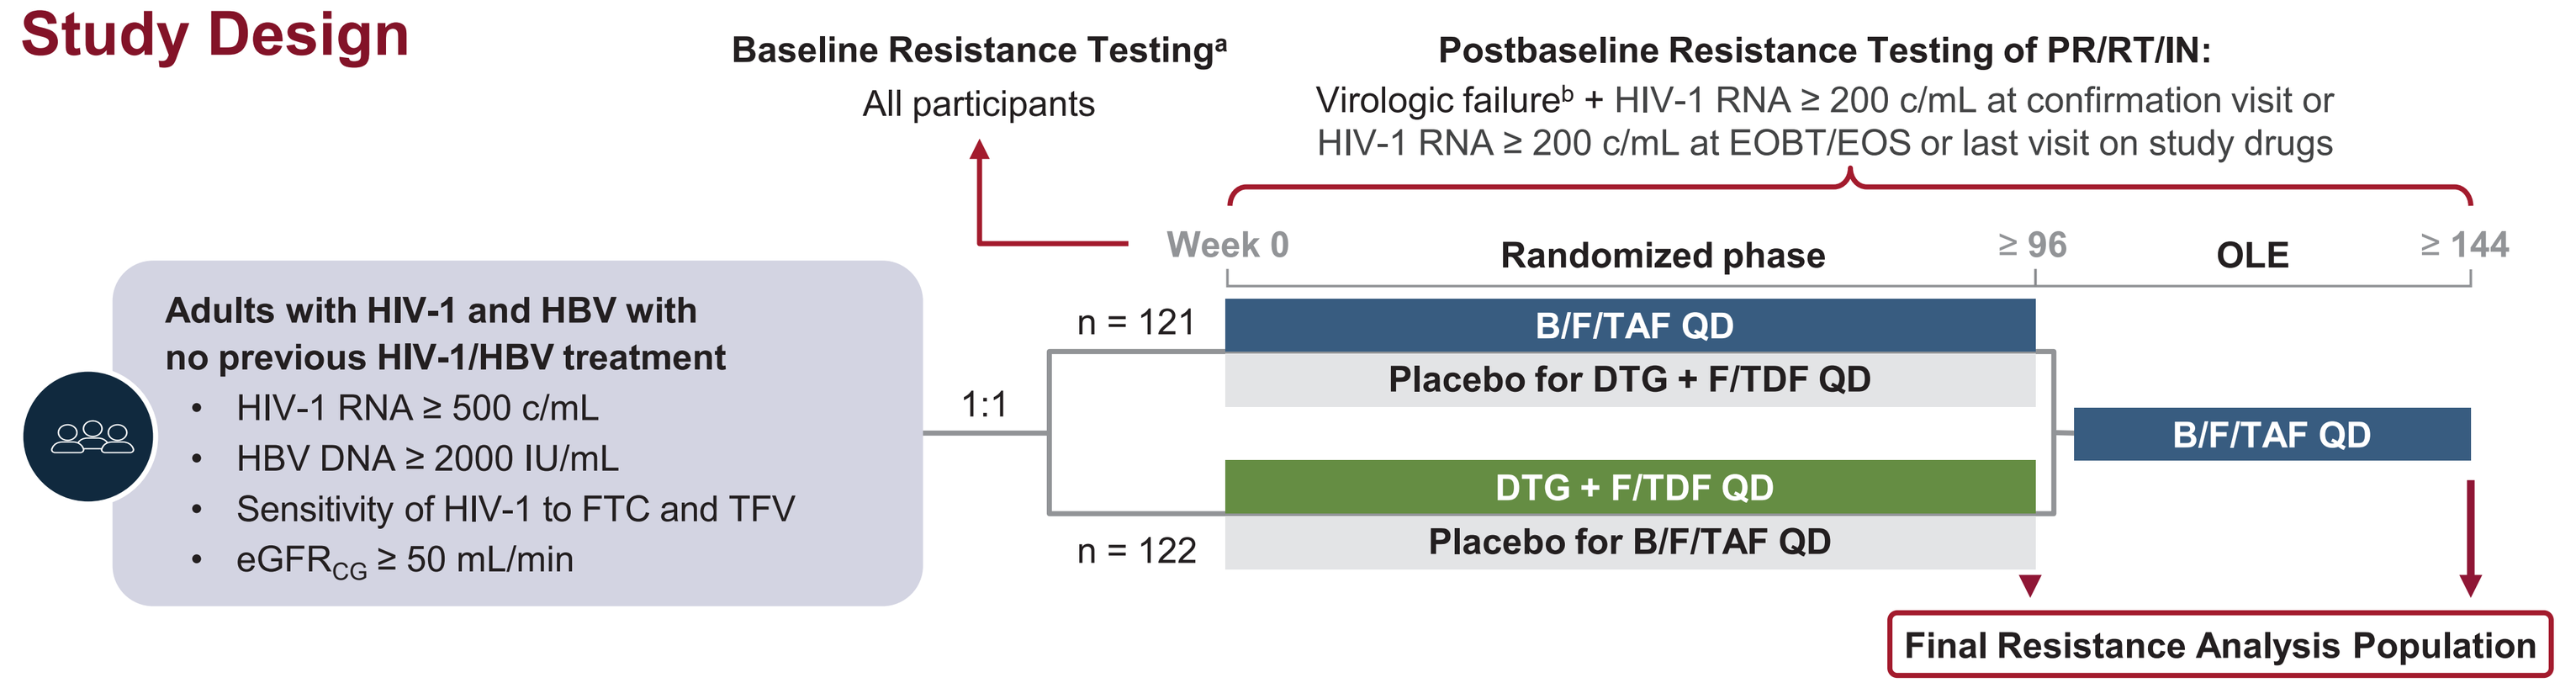
Baseline Resistance Testing[a]
All participants
Postbaseline Resistance Testing of PR/RT/IN:
Virologic failure[b] + HIV-1 RNA ≥ 200 c/mL at confirmation visit or HIV-1 RNA ≥ 200 c/mL at EOBT/EOS or last visit on study drugs
Adults with HIV-1 and HBV with no previous HIV-1/HBV treatment
HIV-1 RNA ≥ 500 c/mL
HBV DNA ≥ 2000 IU/mL
Sensitivity of HIV-1 to FTC and TFV
eGFRCG ≥ 50 mL/min
1:1
Week 0
Randomized phase
≥ 96
OLE
≥ 144
n = 121
B/F/TAF QD
Placebo for DTG + F/TDF QD
n = 122
DTG + F/TDF QD
Placebo for B/F/TAF QD
B/F/TAF QD
Final Resistance Analysis Population

[a]Historical genotypes (PR/RT/IN) were collected if available, and PR/RT genotyping from screening plasma samples was aggregated to generate a composite baseline sequence. [b]Virologic failure was defined as virologic rebound (HIV-1 RNA ≥ 50 c/mL after achieving < 50 c/mL, subsequently confirmed at the following visit; > 1 $\log_{10}$ increase from nadir, subsequently confirmed at the following visit) or HIV-1 RNA ≥ 50 c/mL at last on-treatment visit.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c, copies; DTG, dolutegravir; $eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; EOBT, end of blinded treatment; EOS, end of study; F/TDF, emtricitabine/tenofovir disoproxil fumarate; FTC, emtricitabine; HBV, hepatitis B virus; IN, integrase; OLE, open-label extension; PR, protease; QD, once daily; RT, reverse transcriptase; TFV, tenofovir.

- Participants meeting the criteria for postbaseline resistance testing for protease (PR), reverse transcriptase (RT), and integrase (IN) who did not subsequently achieve resuppression (HIV-1 RNA < 50 copies [c]/mL) on study drugs were included in the final resistance analysis population (RAP) according to treatment group:
 1. **DTG + F/TDF group**: Participants who received DTG + F/TDF in the randomized phase, analyzed through end of blinded treatment
 2. **B/F/TAF group**: Participants who received B/F/TAF in the randomized phase, irrespective of receiving B/F/TAF in the OLE phase, analyzed through end of study (EOS)
 3. **DTG + F/TDF → B/F/TAF group**: Participants who received DTG + F/TDF in the randomized phase and then received ≥ 1 dose of B/F/TAF in the OLE phase, analyzed through EOS
- Virologic outcomes were assessed using the last-observation-carried-forward (LOCF) approach

Results

Baseline Demographic and Clinical Characteristics[5]

	DTG + F/TDF n = 122	B/F/TAF n = 121	DTG + F/TDF → B/F/TAF n = 89[a]
Age, years, median (Q1, Q3)	32 (25, 38)	31 (27, 39)	34 (28, 39)
Male sex at birth, n (%)	120 (98)	112 (93)	87 (98)
Asian race, n (%)	106 (87)	108 (89)	84 (94)
BMI, kg/m^2, median (Q1, Q3)	21.7 (19.3, 23.7)	22.2 (19.9, 24.7)	22.4 (20.4, 25.5)
$eGFR_{CG}$, mL/min, median (Q1, Q3)	104.7 (93.0, 124.2)	106.8 (94.8, 130.8)	94.2 (80.4, 112.8)
HIV-1 RNA, $\log_{10}$ c/mL, median (Q1, Q3)	4.69 (4.26, 5.04)	4.66 (4.22, 5.12)	1.28 (1.28, 1.28)
HIV-1 RNA > 100,000 c/mL, n (%)	34 (28)	38 (31)	0 (0)
HIV-1 subtype B, n (%)	33 (27)	24 (20)	11 (12)
HIV-1 non-B subtype, n (%)	59 (48)[b]	70 (59)[c]	49 (55)[d]
CD4 count, cells/μL, median (Q1, Q3)	236 (121, 380)	245 (127, 383)	497 (320, 617)
HIV-1 disease status: asymptomatic, n (%)	81 (66)	83 (69)	57 (64)
HBV DNA ≥ 8 $\log_{10}$ IU/mL, n (%)	66 (54)	60 (50)	0 (0)

[a]Data for this group refer to the baseline at entry to the OLE phase.
[b]Non-B subtypes: AB (3%), AE (35%), BF (2%), G (2%), complex (7%); there were 30 participants (25%) with unknown HIV-1 subtype.
[c]n = 119; non-B subtypes: A (1%), AB (1%), AE (39%), AG (3%), BC (1%), C (1%), G (3%), complex (10%); there were 25 participants (21%) with unknown HIV-1 subtype.
[d]Non-B subtypes: AB (3%), AE (45%), G (2%), complex (4%); there were 29 participants (33%) with unknown HIV-1 subtype.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BMI, body mass index; c, copies; CD4, cluster of differentiation 4; DTG, dolutegravir; $eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; F/TDF, emtricitabine/tenofovir disoproxil fumarate; HBV, hepatitis B virus; OLE, open-label extension; Q, quartile.

- The presence of primary RAMs affecting nucleoside/nucleotide reverse transcriptase inhibitors (NRTIs) or non-nucleoside reverse transcriptase inhibitors (NNRTIs) had no significant effect on efficacy outcomes
 - NRTI primary RAMs: DTG + F/TDF n = 2, B/F/TAF n = 2, DTG + F/TDF → B/F/TAF n = 2
 - NNRTI primary RAMs: DTG + F/TDF n = 7, B/F/TAF n = 12, DTG + F/TDF → B/F/TAF n = 7

LOCF Efficacy by Baseline NRTI-R

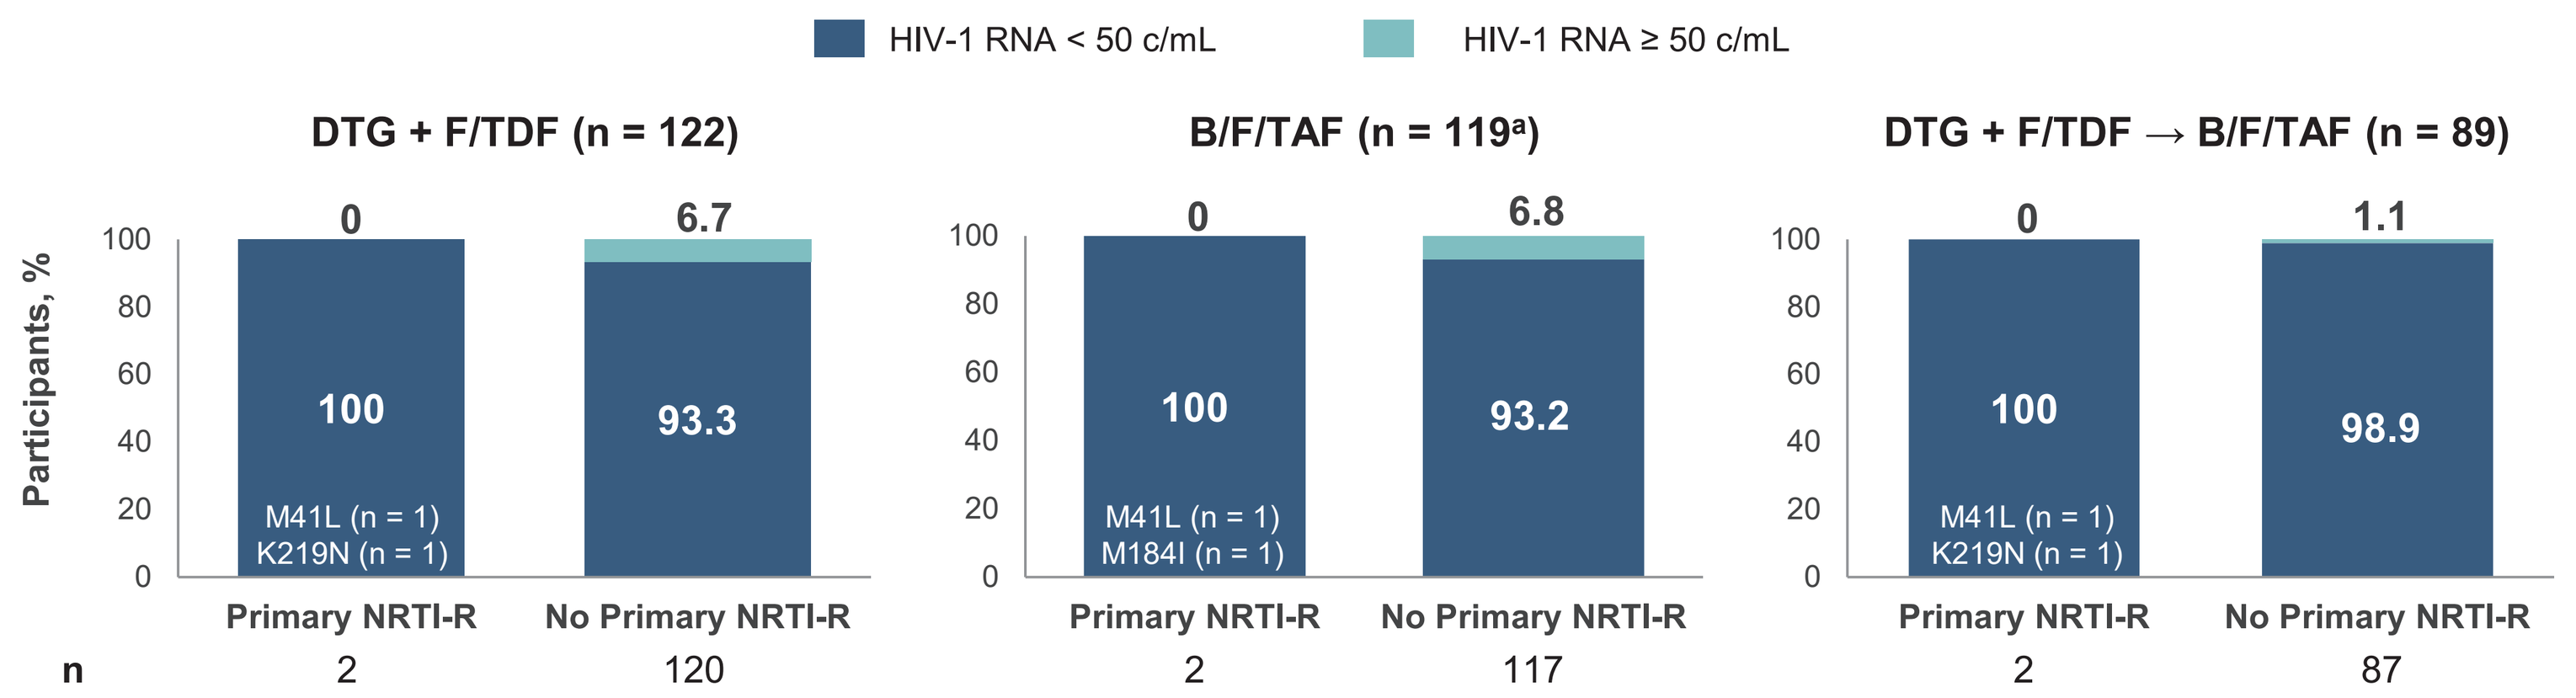
HIV-1 RNA < 50 c/mL
HIV-1 RNA ≥ 50 c/mL
Participants, %
DTG + F/TDF (n = 122)
Primary NRTI-R: 100 / 0; M41L (n = 1), K219N (n = 1); n 2
No Primary NRTI-R: 93.3 / 6.7; n 120
B/F/TAF (n = 119[a])
Primary NRTI-R: 100 / 0; M41L (n = 1), M184I (n = 1); n 2
No Primary NRTI-R: 93.2 / 6.8; n 117
DTG + F/TDF → B/F/TAF (n = 89)
Primary NRTI-R: 100 / 0; M41L (n = 1), K219N (n = 1); n 2
No Primary NRTI-R: 98.9 / 1.1; n 87

[a]Two randomized and treated participants were excluded from the full analysis set due to having no postbaseline HIV-1 RNA or HBV DNA results while on study drug.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c, copies; DTG, dolutegravir; F/TDF, emtricitabine/tenofovir disoproxil fumarate; HBV, hepatitis B virus; LOCF, last observation carried forward; NRTI, nucleoside/nucleotide reverse transcriptase inhibitor; R, resistance.

LOCF Efficacy by Baseline NNRTI-R

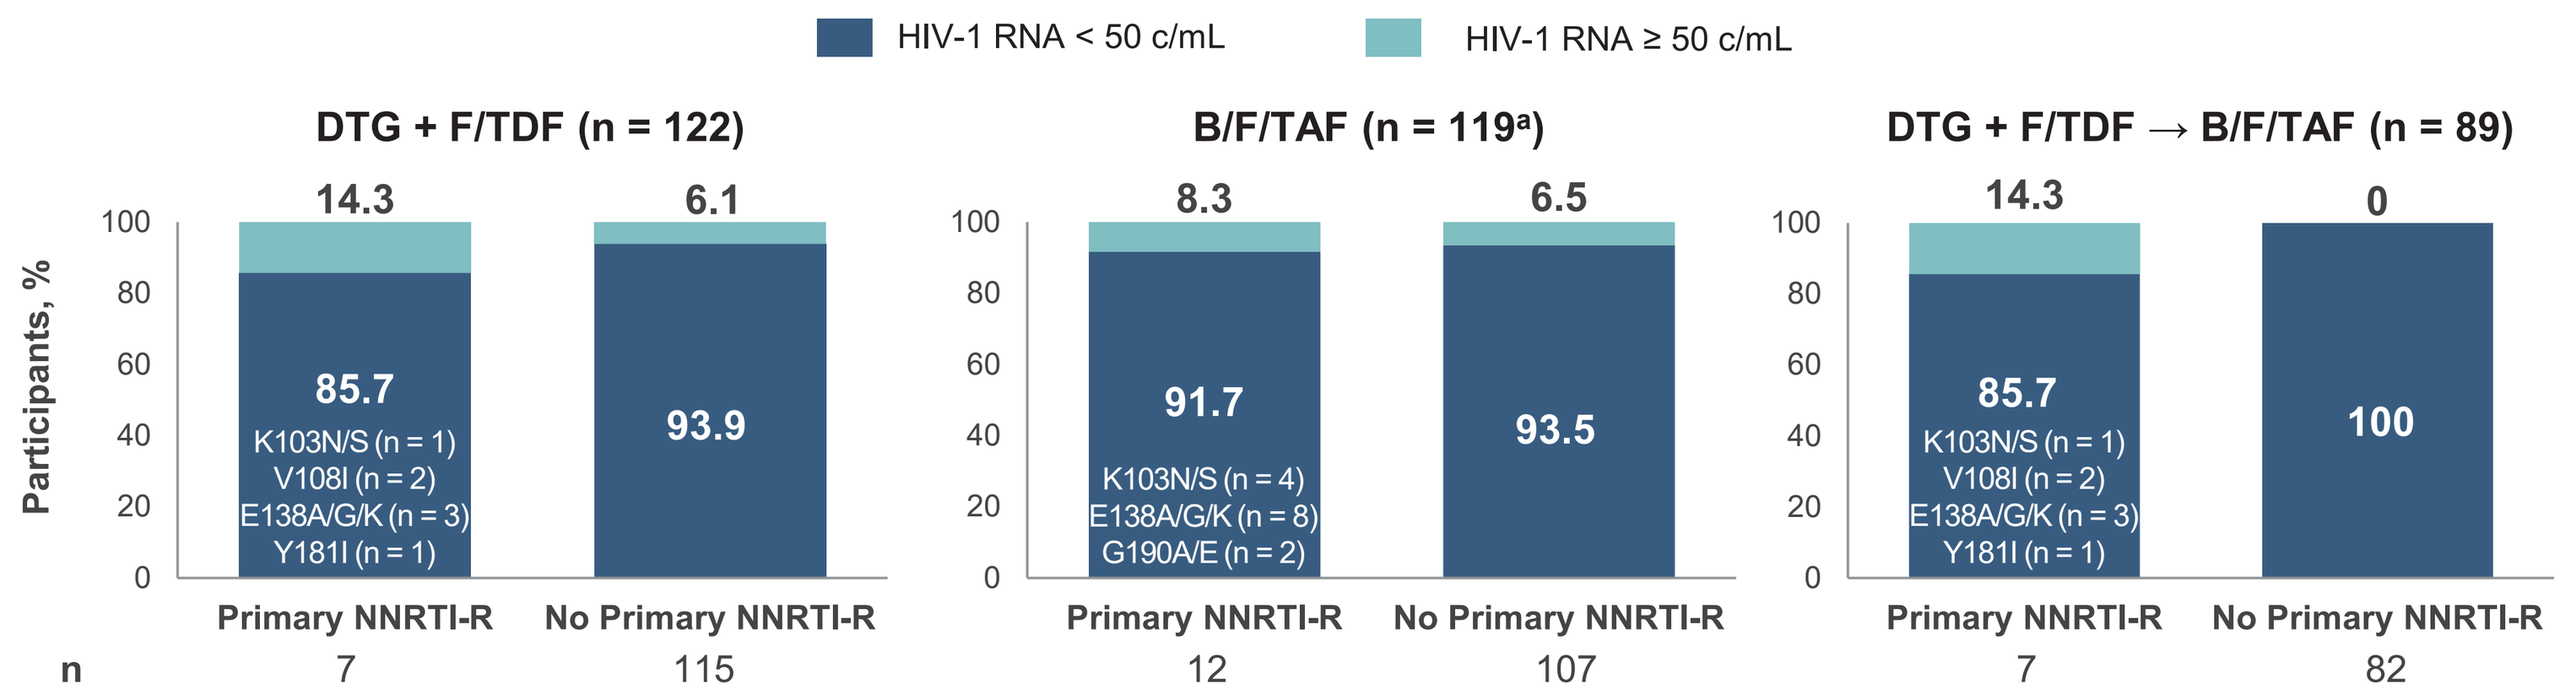
HIV-1 RNA < 50 c/mL
HIV-1 RNA ≥ 50 c/mL
Participants, %
DTG + F/TDF (n = 122)
Primary NNRTI-R: 85.7 / 14.3; K103N/S (n = 1), V108I (n = 2), E138A/G/K (n = 3), Y181I (n = 1); n 7
No Primary NNRTI-R: 93.9 / 6.1; n 115
B/F/TAF (n = 119[a])
Primary NNRTI-R: 91.7 / 8.3; K103N/S (n = 4), E138A/G/K (n = 8), G190A/E (n = 2); n 12
No Primary NNRTI-R: 93.5 / 6.5; n 107
DTG + F/TDF → B/F/TAF (n = 89)
Primary NNRTI-R: 85.7 / 14.3; K103N/S (n = 1), V108I (n = 2), E138A/G/K (n = 3), Y181I (n = 1); n 7
No Primary NNRTI-R: 100 / 0; n 82

[a]Two randomized and treated participants were excluded from the full analysis set due to having no postbaseline HIV-1 RNA or HBV DNA results while on study drug.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c, copies; DTG, dolutegravir; F/TDF, emtricitabine/tenofovir disoproxil fumarate; HBV, hepatitis B virus; LOCF, last observation carried forward; NNRTI, non-nucleoside reverse transcriptase inhibitor; R, resistance.

- Baseline primary RAMs affecting PR inhibitors were rare: DTG + F/TDF n = 4, B/F/TAF n = 1, DTG + F/TDF → B/F/TAF n = 1
 - All of these participants achieved virologic suppression (< 50 c/mL) at EOS
- Seven participants (5.7%) in the DTG + F/TDF group met the criteria for inclusion in the final RAP at the end of the randomized phase
 - No postbaseline primary RAMs were detected in the seven participants with IN data
 - Of the six participants with PR/RT data, none had treatment-emergent primary RAMs, although one had primary NNRTI RAMs at baseline (E138E/G) and at Week 156 (E138G)
 - Emergent secondary RAMs in IN and RT were not associated with genotypic or phenotypic changes to bictegravir, DTG, emtricitabine (FTC), or tenofovir (TFV)
- Six participants (5.0%) in the B/F/TAF group and one participant (1.1%) in the DTG + F/TDF → B/F/TAF group met the criteria for inclusion in the final RAP through EOS
 - Five participants had IN data; none had postbaseline primary RAMs
 - Five participants had postbaseline PR/RT data; none had postbaseline primary RAMs
 - Emergent secondary RAMs in RT were not associated with phenotypic changes to FTC or TFV

HIV-1 Drug RAMs (Based on IAS-USA List)[7]

	Primary RAMs	Secondary RAMs
NRTI-R	K65R/E/N, T69 insertions, K70E, L74V/I, Y115F, Q151M, M184V/I, TAMs (M41L, D67N, K70R, L210W, T215F/Y, K219E/N/Q/R)	E44D, A62V, T69D/N, V75I, F77L, F116Y, V118I, T215A/C/D/E/G/H/I/L/N/S/V
NNRTI-R	L100I, K101E/P, K103N/S, V106A/M, V108I, E138A/G/K/Q/R, V179L, Y181C/I/V, Y188C/H/L, G190A/E/Q/S, H221Y, P225H, F227C, M230I/L	V90I, A98G, K101H, V106I, V179D/F/T
PI-R	D30N, V32I, M46I/L, I47V/A, G48V, I50V/L, I54M/L, Q58E, T74P, L76V, V82A/F/L/S/T, N83D, I84V, N88S, L90M	Not analyzed
INSTI-R	T66I/A/K, E92Q/G, T97A, F121Y, Y143R/H/C, S147G, Q148H/K/R, N155H/S, R263K	M50I, H51Y, L68V/I, V72A/N/T, L74M, Q95K/R, G118R, S119P/R/T, F121C, A128T, E138K/A, G140A/C/S, P145S, Q146R/I/K/L/P, V151L/A, S153A/F/Y, E157K/Q, G163K/R, E170A

IAS-USA, International Antiviral Society–USA; INSTI, integrase strand transfer inhibitor; NNRTI, non-nucleoside reverse transcriptase inhibitor; NRTI, nucleoside/nucleotide reverse transcriptase inhibitor; PI, protease inhibitor; R, resistance; RAM, resistance-associated mutation; TAM, thymidine analog mutation.

Virologic Profiles and RAMs

Participants Receiving DTG + F/TDF in the Final RAP Through EOBT

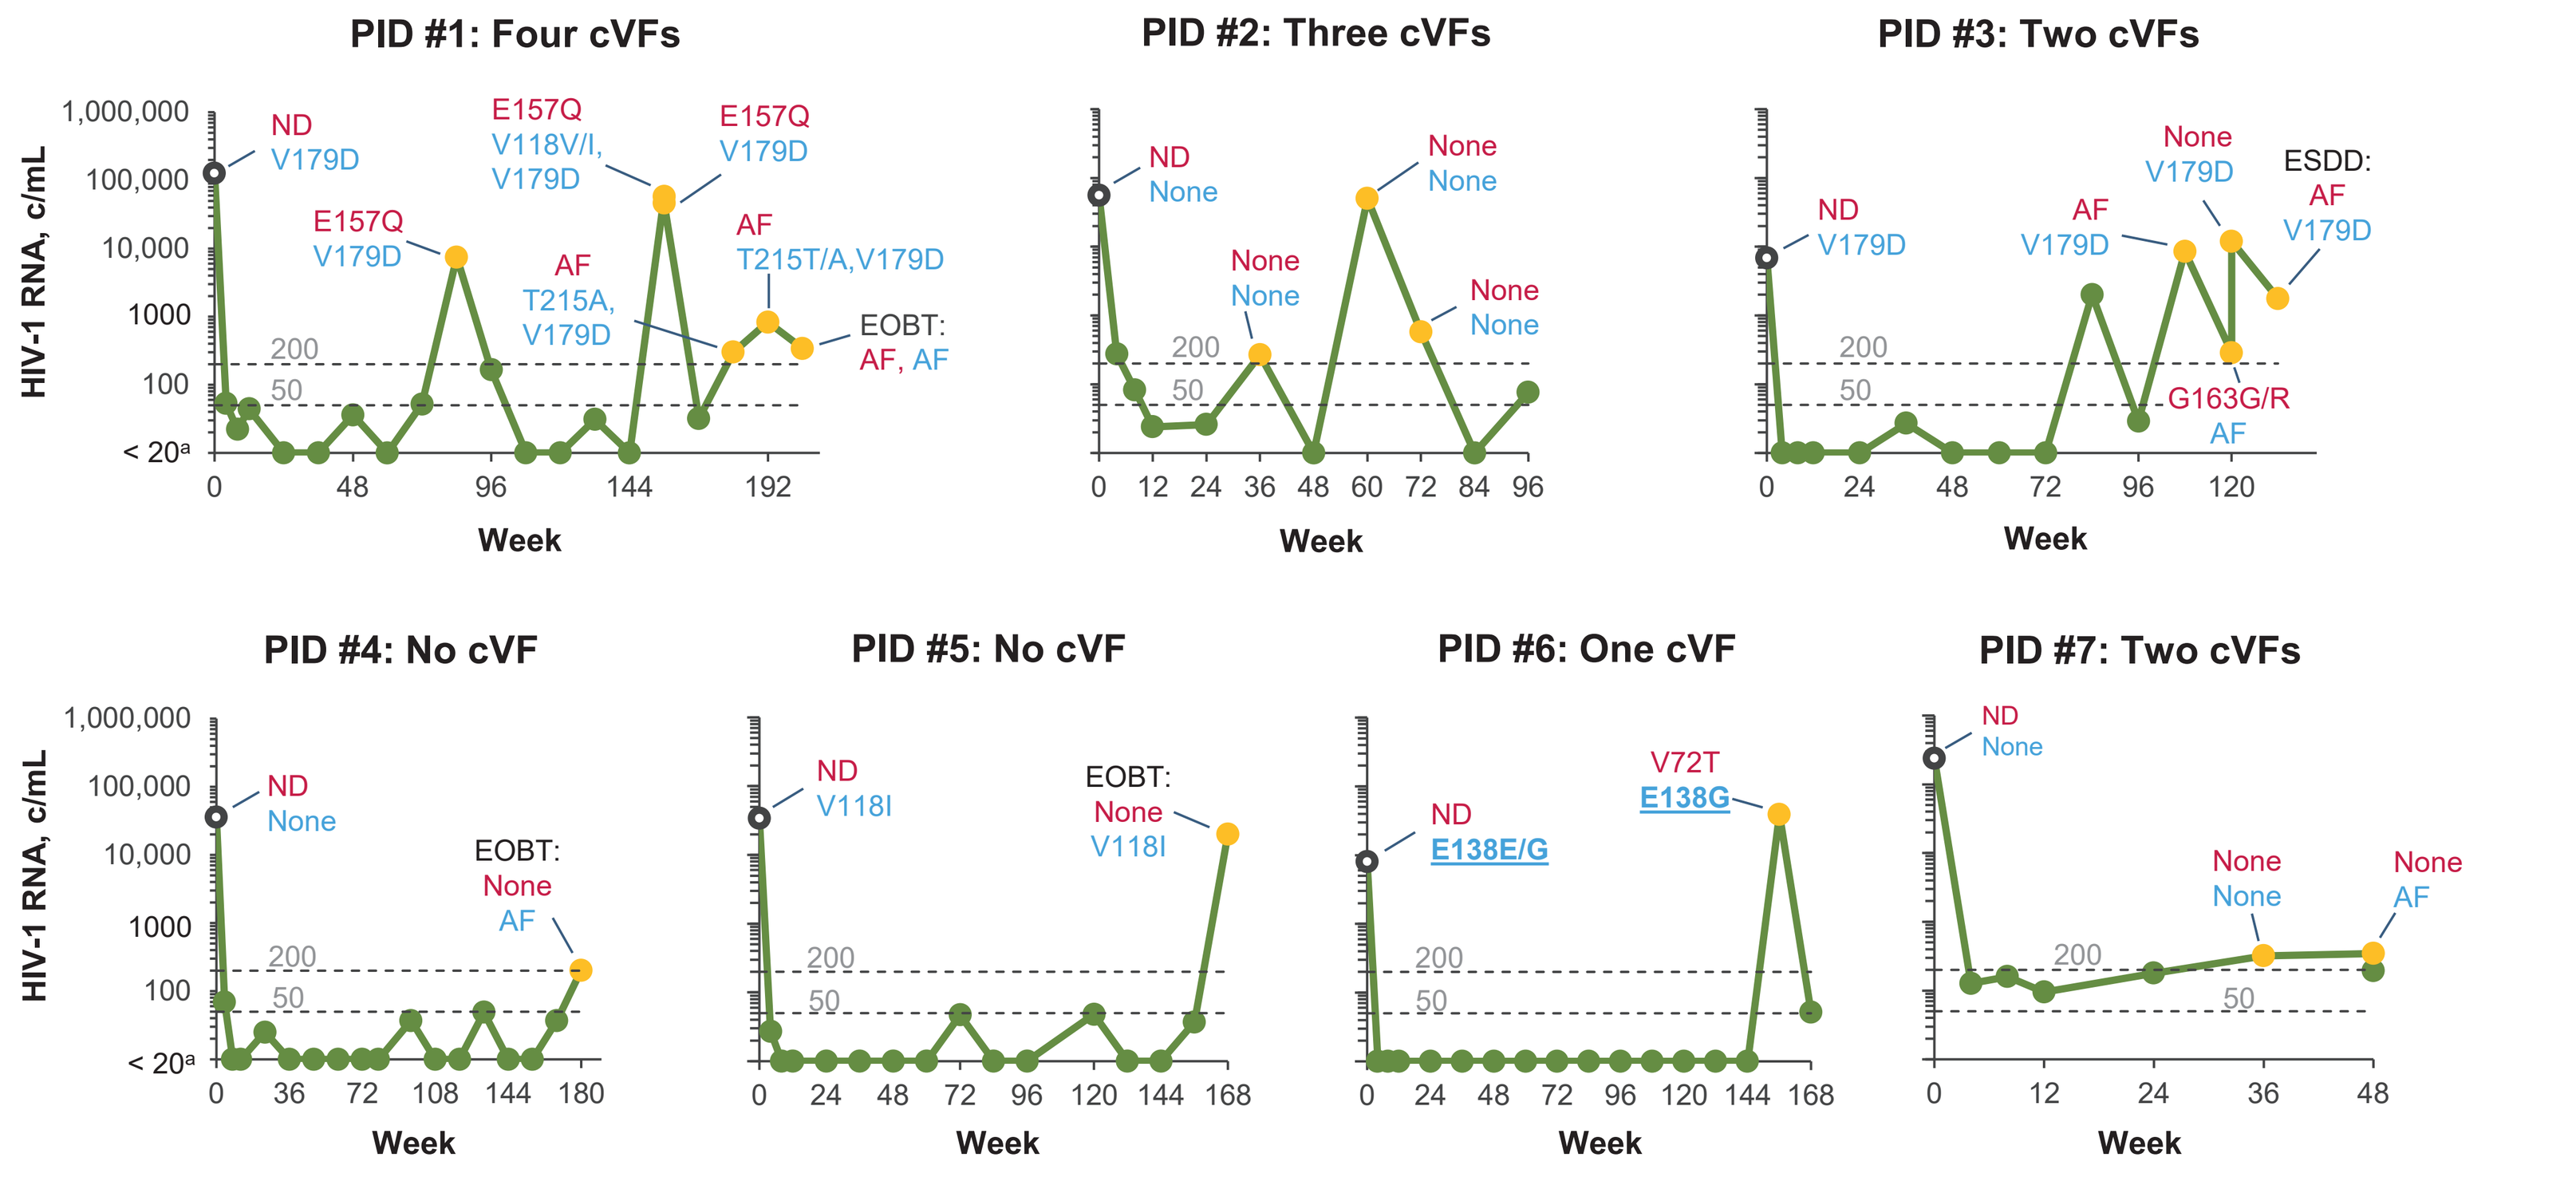
PID #1: Four cVFs
PID #2: Three cVFs
PID #3: Two cVFs
PID #4: No cVF
PID #5: No cVF
PID #6: One cVF
PID #7: Two cVFs
HIV-1 RNA, c/mL
Week

Participants Receiving B/F/TAF in the Final RAP Through EOS

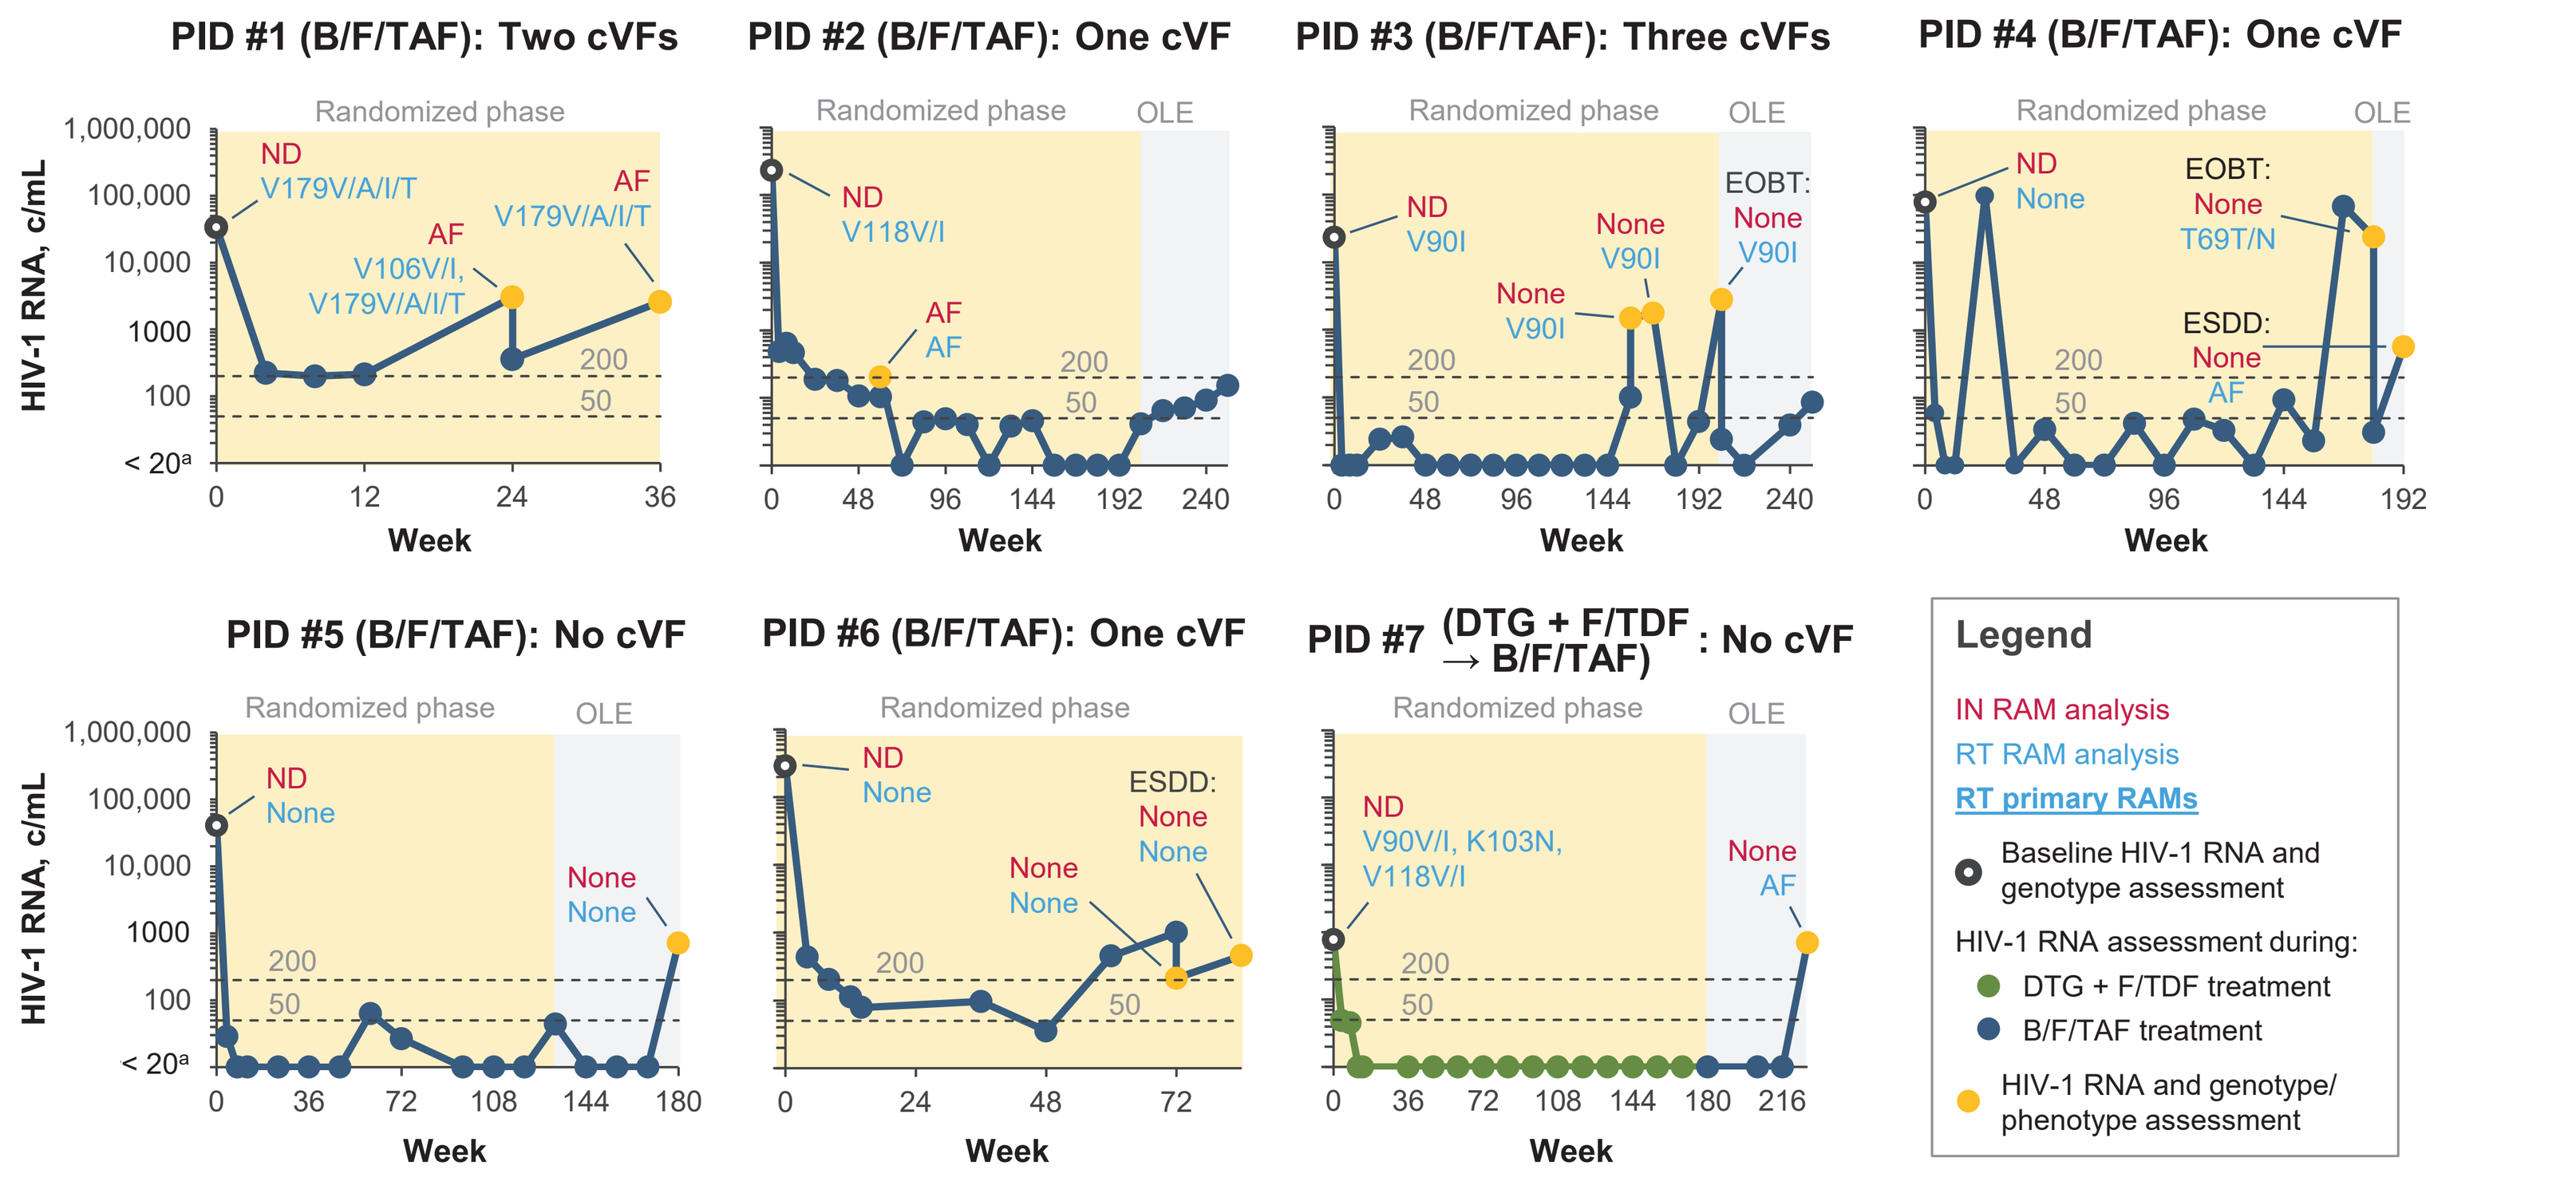
PID #1 (B/F/TAF): Two cVFs
PID #2 (B/F/TAF): One cVF
PID #3 (B/F/TAF): Three cVFs
PID #4 (B/F/TAF): One cVF
PID #5 (B/F/TAF): No cVF
PID #6 (B/F/TAF): One cVF
PID #7 (DTG + F/TDF → B/F/TAF): No cVF
Randomized phase
OLE
HIV-1 RNA, c/mL
Week
Legend
IN RAM analysis
RT RAM analysis
RT primary RAMs
Baseline HIV-1 RNA and genotype assessment
HIV-1 RNA assessment during:
DTG + F/TDF treatment
B/F/TAF treatment
HIV-1 RNA and genotype/phenotype assessment

Week 0 (baseline) sequences are the composite of historical and screening genotyping data.
[a]Includes all readings where HIV-1 RNA was not detected.
AF, assay failure; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c, copies; cVF, confirmed virologic failure; DTG, dolutegravir; EOBT, end of blinded treatment; EOS, end of study; ESDD, early study drug discontinuation; F/TDF, emtricitabine/tenofovir disoproxil fumarate; IN, integrase; ND, no data; OLE, open-label extension; PID, participant ID; RAM, resistance-associated mutation; RAP, resistance analysis population; RT, reverse transcriptase.

Additional Postbaseline Testing

- Another eight participants who did not qualify for the final RAPs had resistance testing during the study due to confirmed virologic failure(s) or having HIV-1 RNA ≥ 200 c/mL at key study endpoints (DTG + F/TDF n = 3, B/F/TAF n = 4, DTG + F/TDF → B/F/TAF n = 1)
- For participants with available IN postbaseline data (n = 5), no INSTI RAMs were detected
- For participants with available PR/RT postbaseline data (n = 4), emergent RAMs (K70E and M184V/I) in RT were detected in one participant receiving DTG + F/TDF
 - This participant had documented nonadherence to DTG + F/TDF throughout the study, experienced three confirmed virologic failures, and subsequently achieved HIV-1 RNA < 50 c/mL on study drugs

References: 1. Leumi S, et al. *Clin Infect Dis.* 2020;71:2799-806. 2. European AIDS Clinical Society. https://www.eacsociety.org/media/guidelines-12.0.pdf (accessed Nov. 18, 2024). 3. Department of Health and Human Services. https://clinicalinfo.hiv.gov/sites/default/files/guidelines/documents/adult-adolescent-arv/guidelines-adult-adolescent-arv.pdf (accessed Nov. 18, 2024). 4. Gandhi RT, et al. *JAMA.* 2023;329:63-84. 5. Avihingsanon A, et al. *Lancet HIV.* 2023;10:e640-52. 6. Nastri BM, et al. *Microorganisms.* 2023;11:221. 7. Wensing AM, et al. *Top Antivir Med.* 2022;30:559-74.

Acknowledgments: We thank all study participants, investigators, and staff. The study was sponsored by Gilead Sciences, Inc. Medical writing support was provided by Noel Curtis, PhD (Aspire Scientific Ltd, UK), and was funded by Gilead Sciences, Inc.

Disclosures: **MLD**, **AVB**, **KA**, **SC**, **JTH**, **LAV**, and **CC** are employees of, and own stocks/shares in, Gilead Sciences, Inc. **AA** reports research/grant support to institute from Gilead Sciences, Inc., GSK/ViiV Healthcare, MSD, and Roche; travel support from Gilead Sciences, Inc.; and membership with the Committee of Thai AIDS Society Strategic, the Strategic and Technical Advisory Group on HIV, Hepatitis and STI for WHO SEARO, and the Thailand National Committee on ART, Hepatitis and TB/TPT.

Correspondence: Michelle L D'Antoni, michelle.dantonibrogan@gilead.com.

Conference on Retroviruses and Opportunistic Infections (CROI); March 9–12, 2025; San Francisco, CA, USA